Poster SA127

BHV-2100, a First-in-Class TRPM3 Antagonist for the Treatment of Neuropathic Pain

Joris Vriens, PhD[1]; Jean-Christophe Vanherck, PhD[2]; Arnaud Marchand, PhD[2]; Bruce Car, DVM, PhD, DACVP[3]; Gene Dubowchik, PhD[3]; Reese Caldwell[3]; Volkan Granit, MD, MSc[3]; Lawrence Marcin, PhD[3]; Patrick Chaltin, PhD[2,4]; Thomas Voets, PhD[1,5]

[1]Laboratory of Ion Channel Research, KU Leuven, Leuven, Belgium; [2]CISTIM Leuven vzw, Leuven, Belgium; [3]Biohaven, New Haven, CT, USA; [4]Centre for Drug Design and Discovery (CD3), KU Leuven, Leuven, Belgium; [5]VIB Center for Brain & Disease Research, Leuven, Belgium

BACKGROUND

- Over the last 25 years, the identification and characterization of transient receptor potential (TRP) ion channels as receptors of painful stimuli in sensory neurons have led to a better understanding of the biology of nociception, fueling development of TRP channel–based next-generation analgesics that target pain at its origin[1-4]
- TRPV1, TRPA1, and TRPM8 have been the most extensively studied TRP channel targets for the development of novel analgesics, but this has not yet resulted in new analgesic drugs for human use[3]
- Clinical development of antagonists for TRPV1 and TRPM8 has been hampered by significant effects on thermosensation and thermoregulation. TRPA1 is a challenging drug target because of the channel's complex gating and pharmacology (**Figure 1**)[1,3,5]
- TRPM3 is a calcium-permeable, nonselective cation channel expressed in a large subset of somatosensory neurons, including nociceptors, both in rodents and in humans. Its activation by chemical ligands or noxious heat evokes pain[6-8]
- TRPM3 is functionally upregulated in animal models of inflammatory hyperalgesia and chemotherapy-induced neuropathic pain[9,10]
- TRPM3-deficient mice do not develop mechanical or thermal hypersensitivity in various pain models[8,11,12]

Figure 1. TRPM3: A Novel Peripheral Target for Neuropathic Pain[1,3]

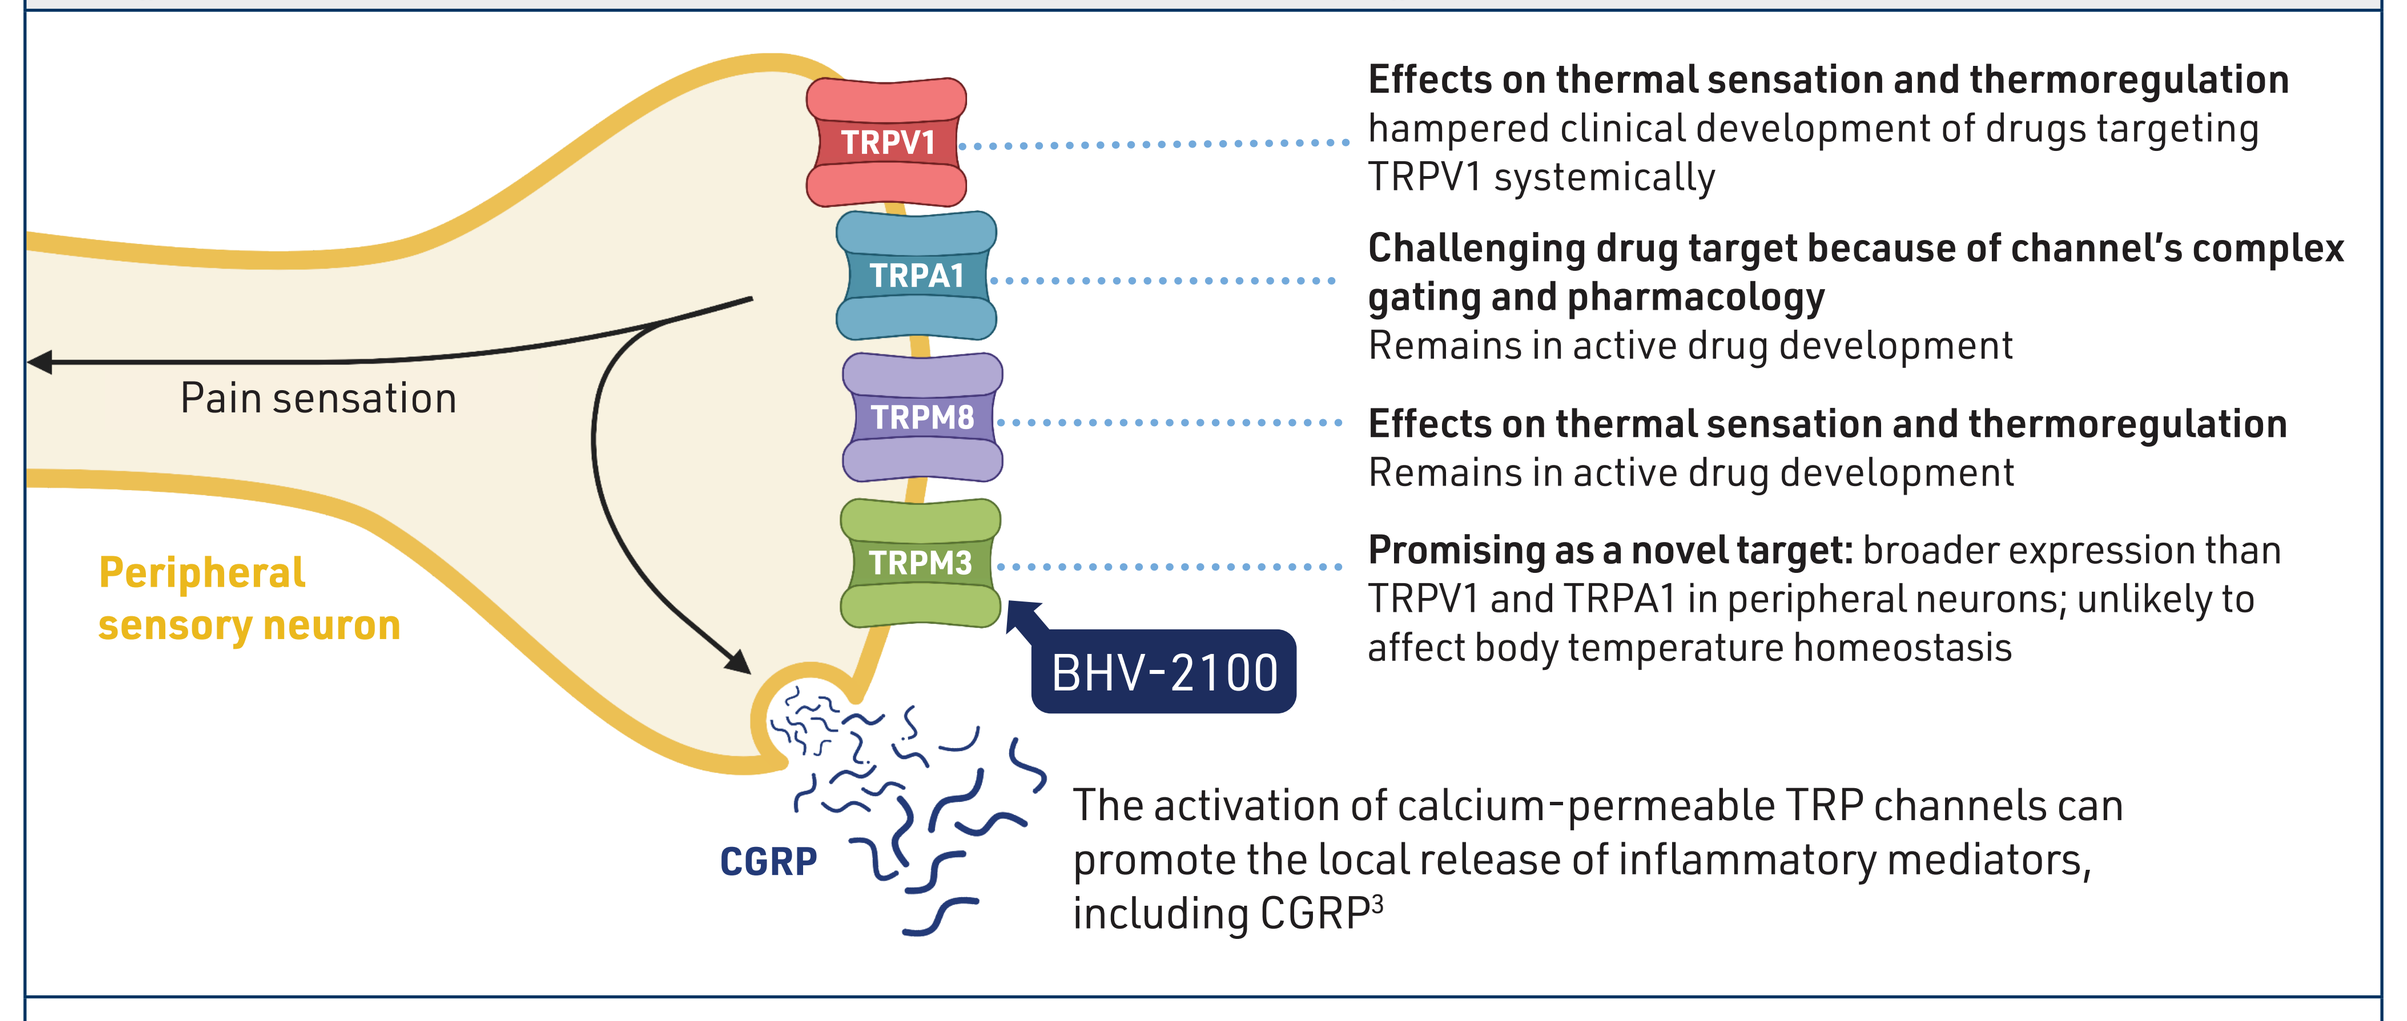
TRPV1
TRPA1
TRPM8
TRPM3
Pain sensation
Peripheral sensory neuron
BHV-2100
CGRP
Effects on thermal sensation and thermoregulation hampered clinical development of drugs targeting TRPV1 systemically
Challenging drug target because of channel's complex gating and pharmacology
Remains in active drug development
Effects on thermal sensation and thermoregulation
Remains in active drug development
Promising as a novel target: broader expression than TRPV1 and TRPA1 in peripheral neurons; unlikely to affect body temperature homeostasis
The activation of calcium-permeable TRP channels can promote the local release of inflammatory mediators, including CGRP3

CGRP, calcitonin gene-related peptide.

- Gain-of-function mutations in TRPM3 have been associated with altered pain sensation in humans. Further, machine learning studies identified single nucleotide polymorphisms in the *TRPM3* gene that are associated with a change in pain thresholds[13,14]
- BHV-2100 is a selective TRPM3 antagonist (**Figure 1**), and we present the key preclinical data supporting its advancement to the clinical stage

METHODS

In Vitro Activity

The in vitro activity of BHV-2100 against TRPM3 and its selectivity toward other relevant ion channels was evaluated using:

- Whole-cell patch-clamp experiments
- Microfluorimetric calcium imaging in transfected HEK293 cells
- Rodent dorsal root ganglion neurons
- Human stem cell–derived sensory neurons

Telemetric Assay

Implantable sensors for longitudinal monitoring of biopotentials evaluated the effect of BHV-2100 in rats on:

- Body core temperature
- Heart rate

Pharmacokinetics and Toxicology

- IND-enabling ADME and toxicology studies were performed

In Vivo Pain Models

The analgesic potential of BHV-2100 was evaluated in a panel of novel and established rodent models, including:

- **Acute pain:** BHV-2100 or vehicle administered 30 minutes prior to intraplantar injection of the TRPM3 agonist pregnenolone sulfate (PS) in the hind paw of mice and rats
- **Nerve injury:** partial sciatic nerve ligation model in rats. BHV-2100, pregabalin, or vehicle administered 14 days after unilateral sciatic nerve injury
- **Chemotherapy-induced neuropathic pain:** mouse oxaliplatin model. BHV-2100, tramadol, or vehicle administered 6 days after oxaliplatin treatment
- **Diabetic neuropathy:** streptozocin-induced diabetic neuropathy model in rats. BHV-2100, pregabalin, or vehicle administered 7 days after streptozocin treatment

ADME, absorption, distribution, metabolism, and excretion; HEK, human embryonic kidney; IND, Investigational New Drug application.

RESULTS

In Vitro Findings

- BHV-2100 inhibited human, mouse, and rat TRPM3-mediated calcium responses in heterologous expression systems and sensory neurons, with half-maximal inhibitory concentration (IC_{50}) values between 1 and 10 nM (**Table 1**)
- BHV-2100 exhibited > 1000-fold selectivity to a large panel of other ion channels and receptors (**Table 1**)

Pharmacokinetics and Toxicology

- BHV-2100 exhibited high oral bioavailability in mice and rats without noticeable side effects (**Table 2**) or impact on body core temperature regulation or heart rate (**Figures 2A and B**), allowing further testing of its efficacy in a panel of in vivo pain models
- Tissue distribution of BHV-2100 activity is highly restricted to the peripheral nervous system and therefore is less susceptible to potential adverse events such as sedation (**Figure 2C**) or abuse liability

In Vivo Pain Models

- Orally administered BHV-2100 inhibited PS-induced pain in rodents with median effective dose (ED_{50}) values of 1.3 mg/kg and 2.5 mg/kg in mice and rats, respectively (**Figure 3A**)
- BHV-2100 also reduced pain in a dose-dependent manner following nerve injury in rats (**Figure 3B**) without the significant sedation effects seen with pregabalin when dosed at 30 mg/kg
- In both the chemotherapy-induced neuropathic pain model and the diabetic neuropathy model, BHV-2100 showed significant dose-dependent efficacy in reversing cold-induced pain and mechanical hypersensitivity, respectively (**Figure 4**)

Table 1. In Vitro Findings

Parameter	Test	Value
TRPM3 electrophysiology	Patch clamp	8.8 nM IC_{50}
TRPM3 neuronal activity	hES-derived sensory neurons	3 nM IC_{50}
TRP selectivity	TRPA1/TRPV1/TRPM8; TRPM7	All > 10 μM IC_{50}
CV selectivity	NaV1.5; NaV1.7; CaV1.2; hERG	All > 10 μM IC_{50}
General selectivity	Eurofins	Clean in BioPrint™

CV, conduction velocity; hERG, human ether-a-go-go-related gene; hES, human embryonic stem cell.

Table 2. Pharmacokinetics and Toxicology Findings

Parameter	Test	Value
ADME	Clearance across species	Low/moderate
ADME	CYP450	All isoforms > 10 μM
ADME	Bioavailability (mouse, rat, dog)	55%-85%
Toxicology	IND-enabling toxicology studies	Wide safety margins, no genotoxicity

CYP450, cytochrome P450.

Figure 2. BHV-2100 Did Not Demonstrate a Significant Impact on BCT, Heart Rate, or Activity

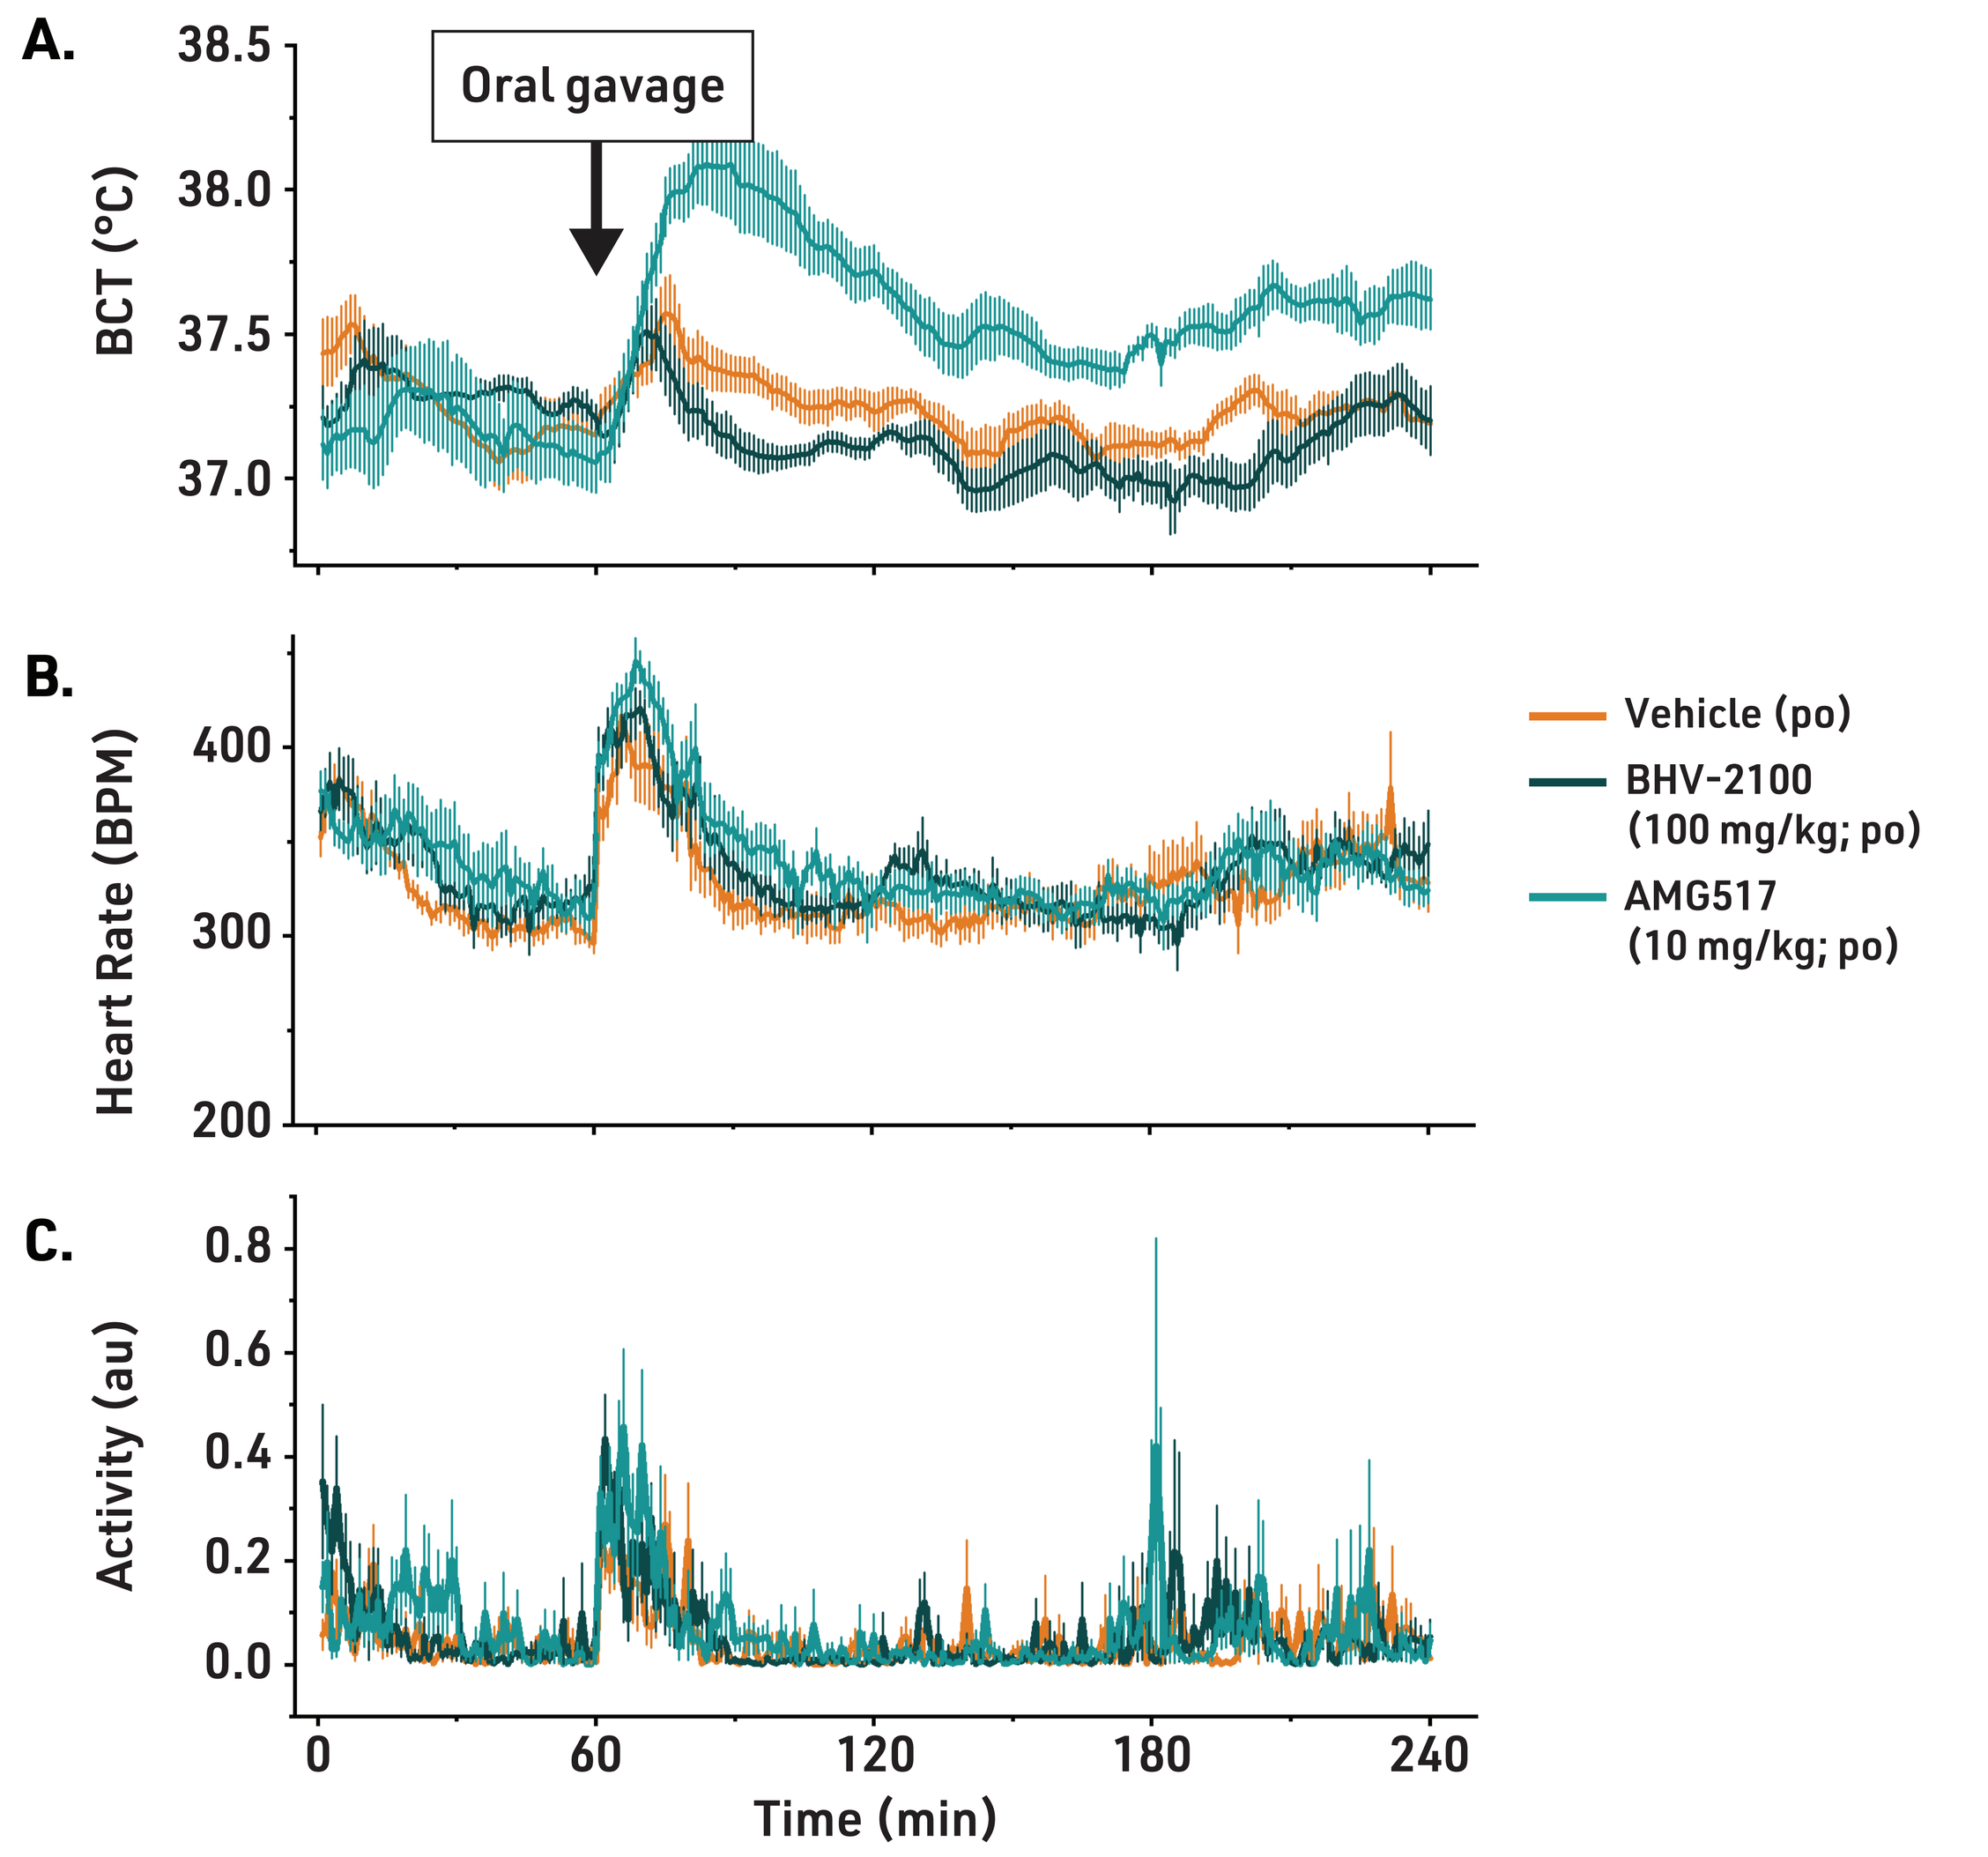
A.
BCT (°C)
38.5
38.0
37.5
37.0
Oral gavage
B.
Heart Rate (BPM)
400
300
200
C.
Activity (au)
0.8
0.6
0.4
0.2
0.0
0
60
120
180
240
Time (min)
Vehicle (po)
BHV-2100 (100 mg/kg; po)
AMG517 (10 mg/kg; po)

Time course of the changes in BCT, activity, and heart rate in rats (n = 8) during 1 hour prior and 3 hours post oral dosing of vehicle, BHV-2100 (100 mg/kg), or AMG517 (10 mg/kg). The parameter activity is expressed in arbitrary units (au), corresponding to the number of automatically detected activity counts per second. AMG517 is a TRPV1 antagonist, causing hyperthermia.[3]
BCT, body core temperature; BPM, beats per minute; po, by mouth.

Figure 3. BHV-2100 Potently Reduces Acute Chemogenic Pain and Pain Following Nerve Injury

A. Pregnenolone Sulfate–Induced Acute Pain Model

B. Partial Sciatic Nerve Ligation Model

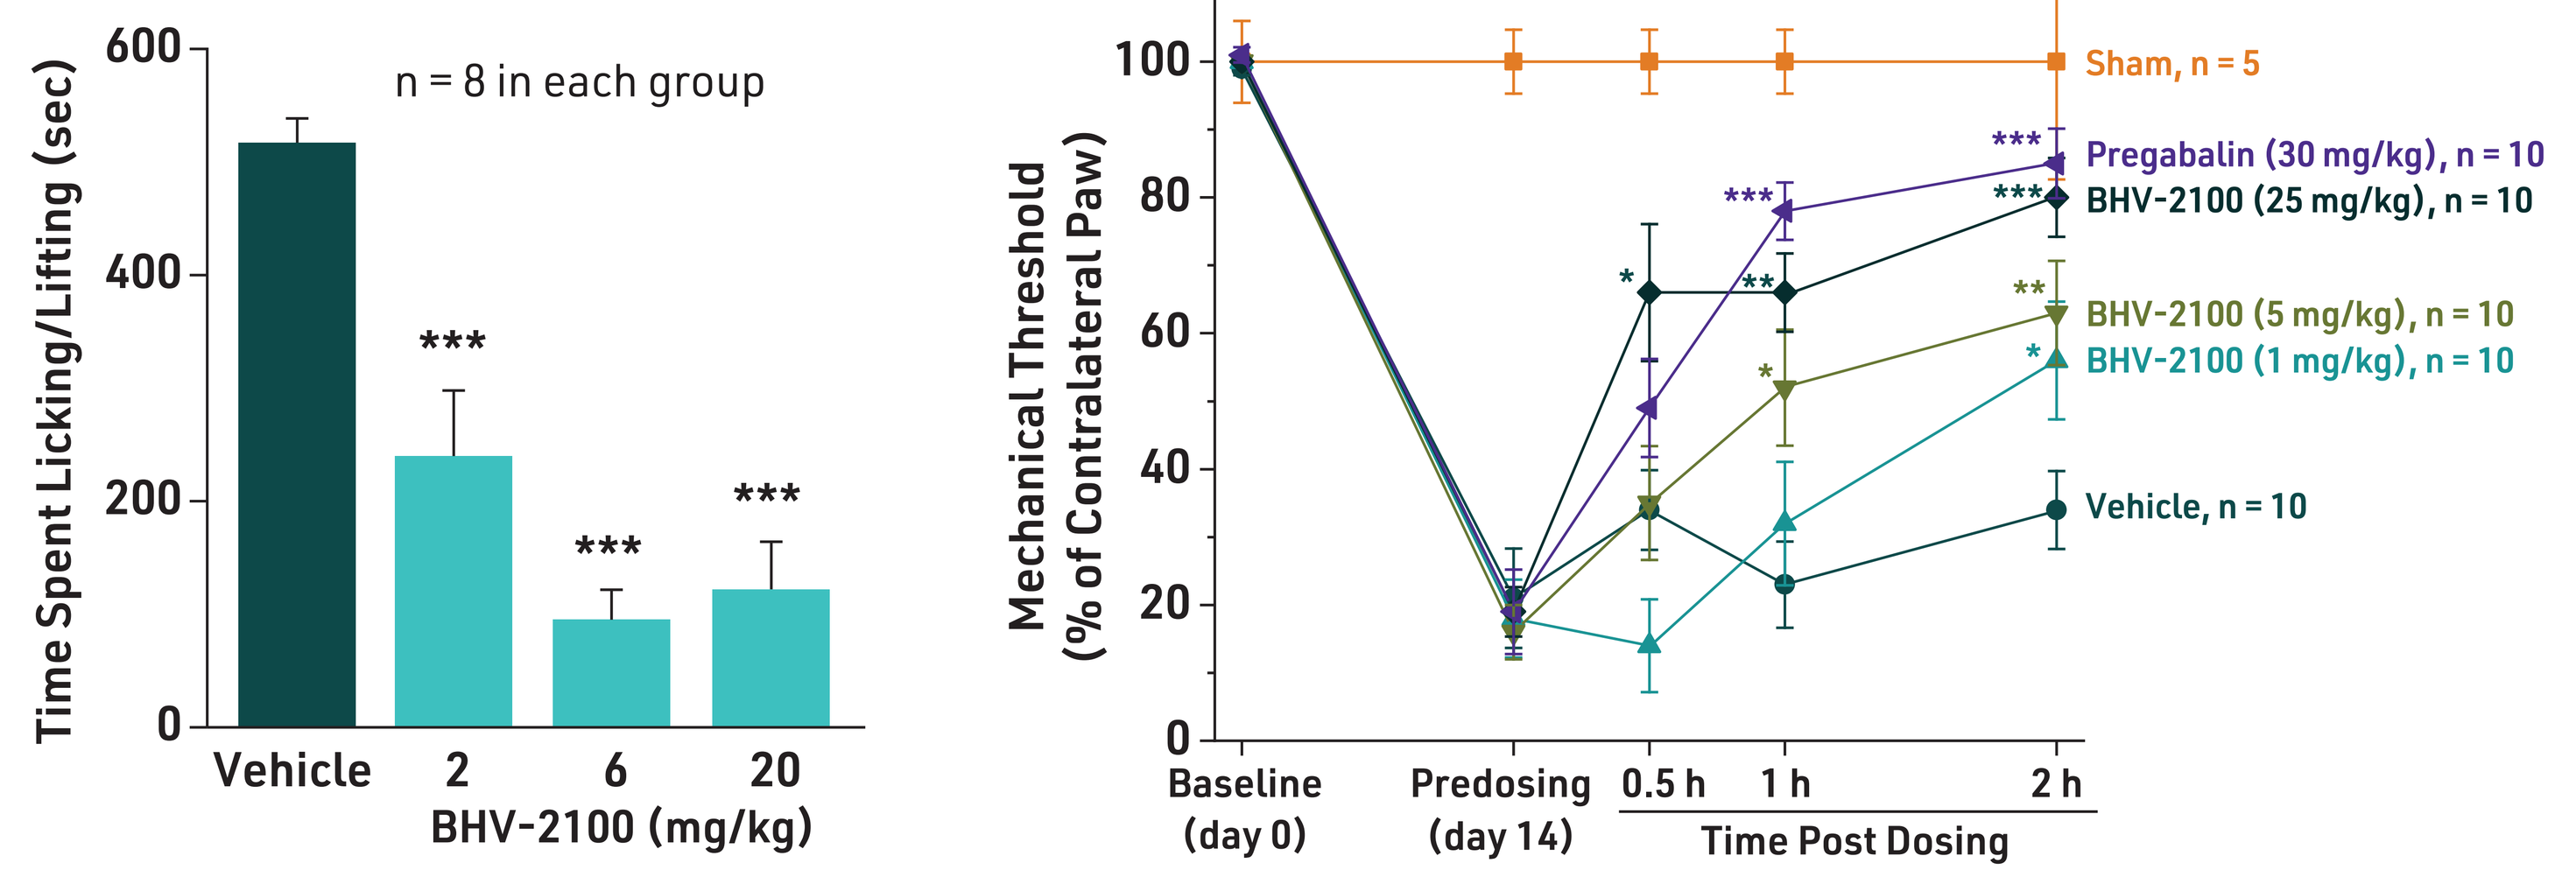
Time Spent Licking/Lifting (sec)
600
400
200
0
n = 8 in each group

Vehicle
2
6
20
BHV-2100 (mg/kg)
Mechanical Threshold (% of Contralateral Paw)
100
80
60
40
20
0
Sham, n = 5
Pregabalin (30 mg/kg), n = 10
BHV-2100 (25 mg/kg), n = 10
BHV-2100 (5 mg/kg), n = 10
BHV-2100 (1 mg/kg), n = 10
Vehicle, n = 10
Baseline (day 0)
Predosing (day 14)
0.5 h
1 h
2 h
Time Post Dosing

Drug administered 30 minutes prior to TRPM3 agonist injection in a hind paw of rats

Drug administered 14 days after unilateral sciatic nerve injury in rats

*$P < 0.05$. **$P < 0.01$. ***$P < 0.001$.

Figure 4. BHV-2100 Reverses Established Pain States in Peripheral Neuropathic Pain Models

A. Chemotherapy-Induced Neuropathic Pain Model

B. Diabetic Neuropathy Model

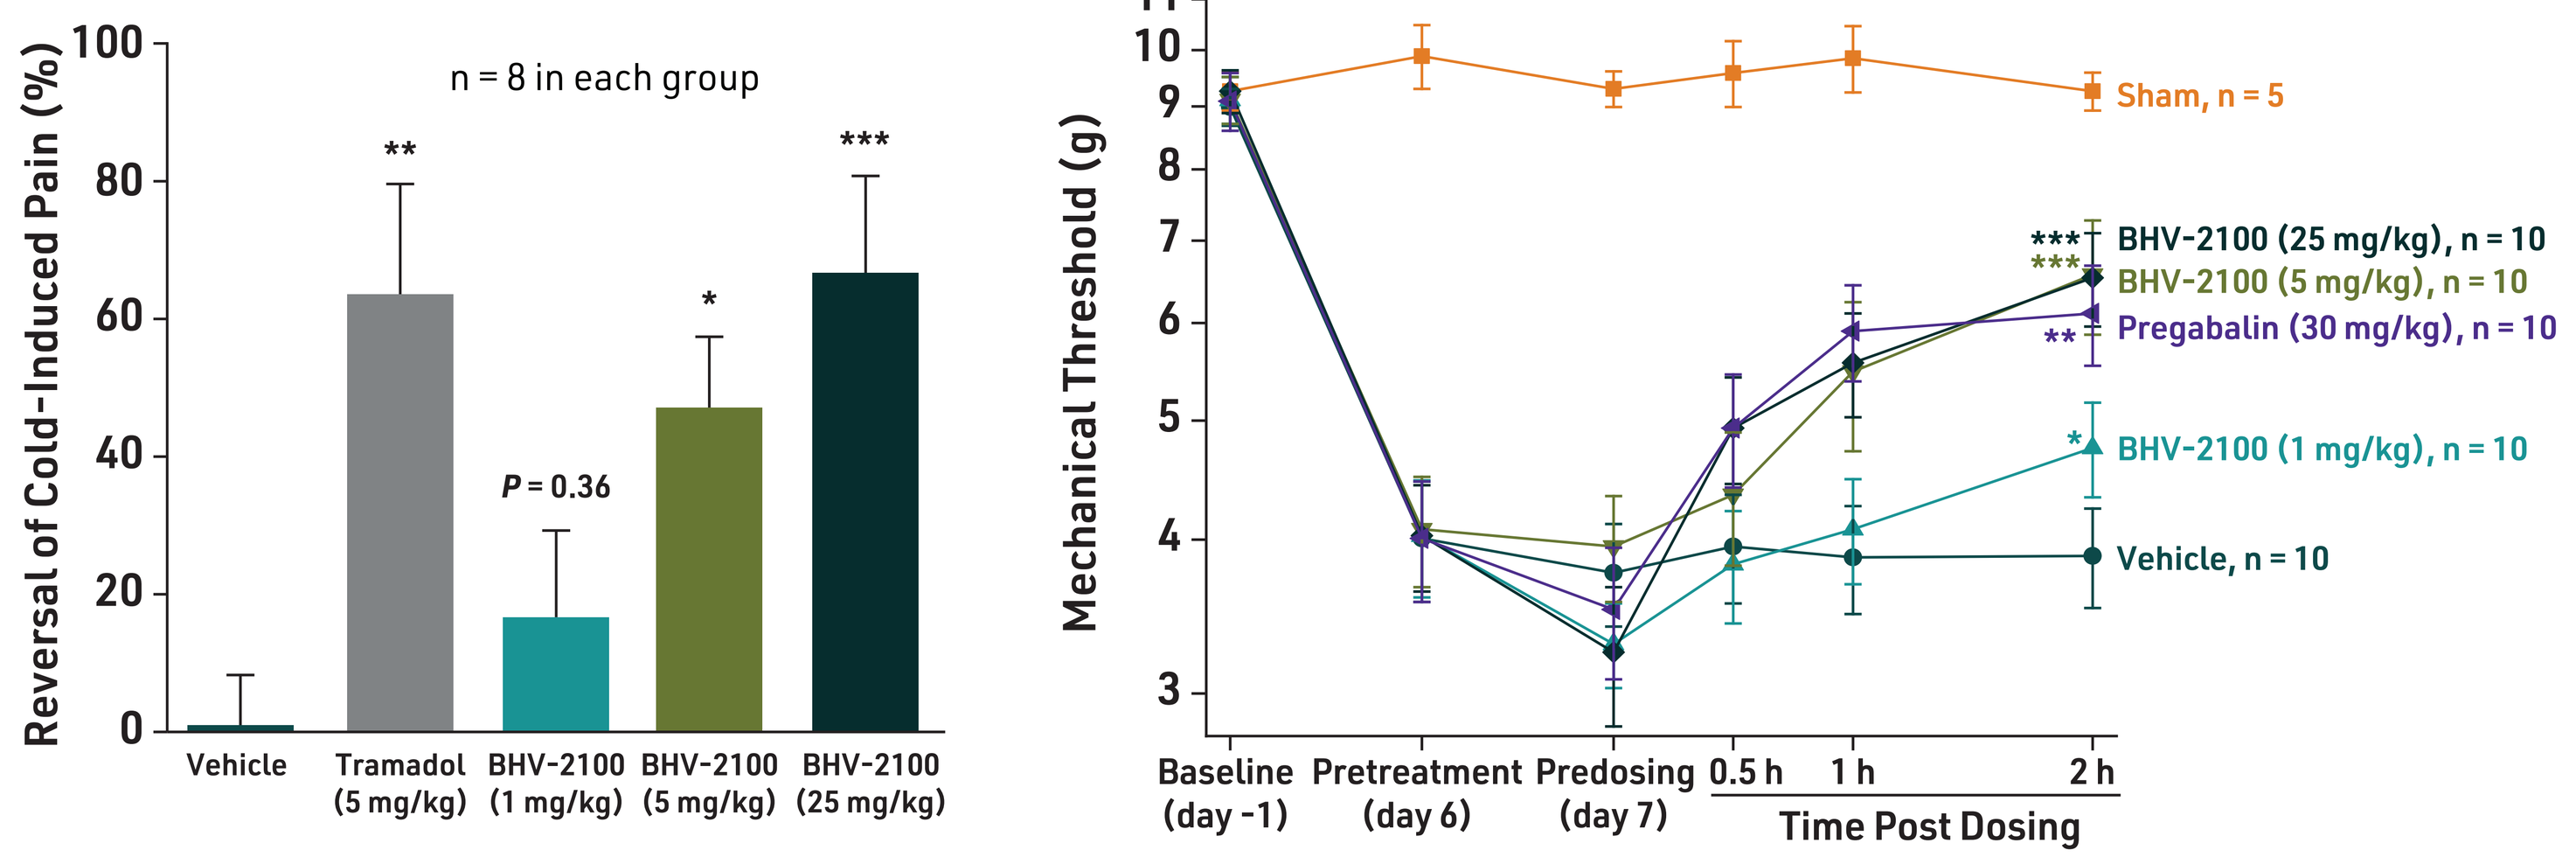
Reversal of Cold-Induced Pain (%)
100
80
60
40
20
0
n = 8 in each group
**
P = 0.36
*

Vehicle
Tramadol (5 mg/kg)
BHV-2100 (1 mg/kg)
BHV-2100 (5 mg/kg)
BHV-2100 (25 mg/kg)
Mechanical Threshold (g)
11
10
9
8
7
6
5
4
3
Sham, n = 5
BHV-2100 (25 mg/kg), n = 10
BHV-2100 (5 mg/kg), n = 10
Pregabalin (30 mg/kg), n = 10
BHV-2100 (1 mg/kg), n = 10
Vehicle, n = 10
Baseline (day -1)
Pretreatment (day 6)
Predosing (day 7)
0.5 h
1 h
2 h
Time Post Dosing

Drug administered 6 days after oxaliplatin treatment in mice

Drug administered 7 days after STZ treatment in rats

*$P < 0.05$. **$P < 0.01$. ***$P < 0.001$.
STZ, streptozotocin.

CONCLUSIONS

1. BHV-2100 is a first-in-class, orally administered, highly potent, and selective TRPM3 antagonist
2. Preclinical data show potent reversal of pain with reduced potential for thermoregulatory side effects or sedation
3. The findings support the hypothesis that TRPM3 represents a safe and druggable target as a novel nonopioid treatment of chronic pain
4. Preclinical data support the advancement of BHV-2100 toward first-in-human studies

References

1. Koivisto AP, et al. *Nat Rev Drug Discov*. 2022;21(1):41-59. **2.** Mickle AD, et al. *Pharmaceuticals*. 2016;9(4):72. **3.** Bamps D, et al. *Annu Rev Pharmacol Toxicol*. 2021;61:655-677. **4.** Patapoutian A, et al. *Nat Rev Drug Discov*. 2009;8(1):55-68. **5.** Fernández-Carvajal A, et al. *Expert Opin Investig Drugs*. 2020;29(11):1209-1222. **6.** Vandewauw I, et al. *Nature*. 2018;555(7698):662-666. **7.** Vangeel L, et al. *Br J Pharmacol*. 2020;177(12):2683-2695. **8.** Vriens J, et al. *Neuron*. 2011;70(3):482-494. **9.** Aloi VD, et al. *Pain*. 2023;164(9):2060-2069. **10.** Mulier M, et al. *Elife*. 2020;9:e61103. **11.** Alkhatib O, et al. *J Neurosci*. 2019;39(40):7840-7852. **12.** Su S, et al. *J Neurosci*. 2021;41(11):2457-2474. **13.** Zhao S, et al. *Elife*. 2020;9:e55634. **14.** Lötsch J, et al. *Int J Mol Sci*. 2020;21(12):4367.

Disclosures

BC, GD, VG, and LM are employed by and hold stock/stock options in Biohaven; RC is a former employee of Biohaven. JV, JCV, AM, PC, and TV have nothing to disclose.

Acknowledgments

The authors would like to thank Kanaka Sridharan, MS, Medical Affairs consultant at Biohaven for providing editorial support and Sara Sloan and Shannon Davis of Apollo Medical Communications for layout of the poster. The authors from Leuven would also like to thank Grünenthal for their contribution.

To download a copy of this poster, scan QR code.

IASP Neuropathic Pain Special Interest Group (NeuPSIG) 2023 International Congress on Neuropathic Pain · 7-9 September 2023 · Lisbon, Portugal

biohaven